ICAAC 2011
C2-1271

In Vitro Activities of Ceftaroline Against *Streptococcus pneumoniae* Collected From Across Canada: CANWARD 2009 - 2010

H.J. Adam,[1,2] A.K. Wierzbowski,[1] J.A. Karlowsky,[1,2] D.J. Hoban,[1,2] G.G. Zhanel[1]

University of Manitoba[1] and Diagnostic Services of Manitoba[2], Winnipeg, Canada

Heather Adam
Diagnostic Services of Manitoba
Clinical Microbiology, Health Sciences Centre
MS673-820 Sherbrook St.
Winnipeg, MB R3A 1R9
Canada
Tel: 1-204-787-8678
Fax: 1-204-787-4699
E-mail: hadam@hsc.mb.ca

Abstract

Background: Ceftaroline is a cephalosporin with broad-spectrum in vitro activity that includes methicillin-resistant *Staphylococcus aureus* (MRSA) and penicillin (PEN)-resistant *Streptococcus pneumoniae* (SPN). The activities of ceftaroline and ceftriaxone against SPN with various resistance phenotypes obtained from Canadian hospitals were assessed between 2009 and 2010, as part of the CANWARD surveillance study.

Methods: Between 2009 and 2010, 420 SPN isolates [218 (51.9%) from blood and 202 (48.1%) from respiratory samples] were submitted from 15 sentinel Canadian hospitals to the CANWARD surveillance study. Susceptibility testing was performed using CLSI broth microdilution. Ceftriaxone and ceftaroline MICs were interpreted with CLSI M100-S21 and FDA breakpoints, respectively. Isolates (n=323) were serotyped by the Quellung method.

Results: The in vitro activities of ceftaroline and ceftriaxone were:

SPN Isolates (n)	%S	MIC_{50} (µg/mL)	MIC_{90} (µg/mL)	Range (µg/mL)
All (420)				
Ceftaroline	100	≤0.03	≤0.03	≤0.03-0.25
Ceftriaxone	99*/98**	≤0.12	≤0.12	≤0.12-4
PEN MIC ≤0.06 µg/mL (362)				
Ceftaroline	100	≤0.03	≤0.03	≤0.03-0.25
Ceftriaxone	100*/100**	≤0.12	≤0.12	≤0.12-0.25
PEN MIC ≥0.12 µg/mL (58)				
Ceftaroline	100	≤0.03	0.12	≤0.03-0.25
Ceftriaxone	93*/84**	≤0.12	1	≤0.12-4
MDNS (32)				
Ceftaroline	100	0.06	0.12	≤0.03-0.25
Ceftriaxone	88*/76**	≤0.12	2	≤0.12-4

* Ceftriaxone nonmeningitis breakpoint.
** Ceftriaxone meningitis breakpoint.
MIC, minimum inhibitory concentration; MDNS, multidrug-nonsusceptible (PEN MIC ≥0.12 µg/mL and clarithromycin MIC >0.5 µg/mL); PEN, penicillin; S, susceptible; SPN, *Streptococcus pneumoniae*.

Forty serotypes were observed among the 323 serotyped isolates. The most common serotypes were 19A (n=44), 22F (n=28), 3 (n=24), 4 (n=19), 7F (n=18), and 23A (n=18). Eleven serotypes were observed among the MDNS (n=31), most commonly 19A (n=12). The ceftaroline MIC_{50}/MIC_{90} for 19A strains was ≤0.03/0.12 µg/mL.

Conclusions: Ceftaroline demonstrated more potent in vitro activity than ceftriaxone (by 8- to 16-fold). Susceptibility rates for ceftaroline were higher than for ceftriaxone against PEN-nonsusceptible and MDNS bacteremic and respiratory isolates of SPN isolated from patients in Canada. Ceftaroline remains highly active against emerging resistant SPN strains, such as 19A.

Introduction

Streptococcus pneumoniae is an important pathogen causing respiratory tract infections such as community-acquired pneumonia and otitis media as well as invasive infections such as bacteremia and meningitis. *S. pneumoniae* is the third most commonly isolated pathogen overall in the Canadian Ward Surveillance (CANWARD) study, an ongoing, national surveillance study assessing the prevalence of pathogens and their antimicrobial resistance patterns in Canadian hospitals (1). *S. pneumoniae* is the most common respiratory pathogen and the fifth most common organism isolated from blood cultures in the CANWARD study (1).

β-lactams have been the mainstay of therapy for *S. pneumoniae* infections. Rising penicillin-nonsusceptibility rates in *S. pneumoniae* have led to increasing usage of macrolides and fluoroquinolones. In addition to the impact of increasing antimicrobial usage, the successful implementation of the 7-valent pneumococcal conjugate vaccine has altered the epidemiology of *S. pneumoniae*. The emergence of multidrug-resistant (MDR) serotype 19A is of considerable public health concern. As a result, the 13-valent pneumococcal conjugate vaccine, which includes serotype 19A, has been developed.

Additionally, the development of antimicrobial agents with good activity against *S. pneumoniae*, including penicillin- and MDR strains, is essential. Ceftaroline (the active form of ceftaroline fosamil) is a cephalosporin with broad-spectrum in vitro activity that includes methicillin-resistant *Staphylococcus aureus* (MRSA) and penicillin-resistant *S. pneumoniae* (2). Ceftaroline has a high affinity for altered penicillin-binding proteins (PBPs) such as PBP2a of MRSA and PBP2x of *S. pneumoniae*, resulting in greater activity than the older β-lactams (3). High clinical cure rates and microbiological response rates were recently reported for hospitalized patients with community-acquired bacterial pneumonia who were treated with ceftaroline fosamil, the prodrug of ceftaroline (4). Ceftaroline fosamil (Teflaro®, Forest Laboratories, Inc.) was approved by the US Food and Drug Administration (FDA) in 2010 for treatment of acute bacterial skin and skin structure infections and community-acquired bacterial pneumonia.

The purpose of this study was to evaluate the in vitro activities of ceftaroline and ceftriaxone against *S. pneumoniae* isolated from blood culture and respiratory tract specimens from across Canada, as part of CANWARD. In particular, the in vitro activities of ceftaroline and ceftriaxone against penicillin- and multidrug-nonsusceptible *S. pneumoniae* and the current predominating serotypes were studied.

Methods

Details of the CANWARD study are available at www.can-r.ca and are described in Zhanel et al. (1).

Bacterial Isolates

Tertiary-care centres from all regions of Canada participated in the CANWARD study, including 15 centres in 2009 and 14 centres in 2010. Annually, each centre submitted pathogens (consecutive, 1 per patient/infection site) from blood, respiratory, urinary tract, and wound infections from patients attending hospital clinics, emergency rooms, medical and surgical wards, and intensive care units. Specifically, centres were asked to submit 165 blood (15 per month, Jan. – Nov.), 100 respiratory, 50 urine, and 50 wound specimens each year. The organisms were deemed to be clinically significant by the local centre criteria. Organism identification was performed by the submitting site and confirmed at the coordinating laboratory (Health Sciences Centre, Winnipeg, Manitoba) as required, based on morphological characteristics and antimicrobial susceptibility patterns. Isolates were submitted to the coordinating laboratory, subcultured onto appropriate media, and stocked in skim milk at -80°C. A total of 10,244 organisms were collected between 2009 and 2010, including 420 *S. pneumoniae* (218 from blood and 202 from respiratory samples).

Antimicrobial Susceptibility Testing

Antimicrobial susceptibility testing was performed using the broth microdilution method recommended by the Clinical and Laboratory Standards Institute (CLSI) (5). Custom-designed broth microdilution panels were prepared in-house with cation-adjusted Mueller-Hinton broth with 5% laked horse blood and antimicrobial agents obtained as laboratory-grade powders from their respective manufacturers or from Sigma-Aldrich (Oakville, Canada), as required. The concentration ranges (µg/mL) utilized for ceftaroline and ceftriaxone were 0.03 – 16 and 0.12 – 8, respectively. Organisms were subcultured twice from frozen stocks prior to antimicrobial susceptibility testing. Colony counts were performed periodically to confirm inocula. Quality control was performed using *S. pneumoniae* ATCC 49619. Ceftriaxone and ceftaroline MICs were interpreted with CLSI M100-S21 and US FDA breakpoints, respectively.

Serotyping

The serotype of each isolate was determined by the Quellung reaction following standard methodology (6) using type-specific antisera obtained from the Statens Serum Institute (Copenhagen, Denmark). Infrequently observed serotypes were confirmed by the US Centers for Disease Control and Prevention's polymerase chain reaction (PCR) methodology (http://www.cdc.gov/ncidod/biotech/strep/pcr.htm) at the National Microbiology Laboratory – Public Health Agency of Canada. Serotyping of 323 isolates was completed.

Results

Table 1. Demographics of the *Streptococcus pneumoniae* Isolates Collected in CANWARD, 2009 – 2010

	All	Blood	Respiratory
	Number of isolates (%)		
All	420	218	202
Age (years)			
≤ 17	66 (16)	37 (17)	29 (14)
18-64	194 (46)	102 (47)	92 (46)
≥ 65	160 (38)	79 (36)	81 (40)
Gender			
Female	193 (46)	102 (47)	91 (45)
Male	227 (54)	116 (53)	111 (55)
Geographical region (province)			
British Columbia/ Alberta	57 (14)	33 (15)	24 (12)
Saskatchewan/ Manitoba	79 (19)	33 (15)	46 (23)
Ontario	116 (28)	64 (29)	52 (26)
Quebec	85 (20)	63 (29)	22 (11)
Nova Scotia/ New Brunswick	83 (20)	25 (11)	58 (29)
Hospital location			
Clinics/ offices	65 (15)	10 (5)	55 (27)
Emergency rooms	163 (39)	123 (56)	40 (20)
Intensive care units	77 (18)	19 (9)	58 (29)
Medical wards	89 (21)	52 (24)	37 (18)
Surgical wards	26 (6)	14 (6)	12 (6)

Table 2. In vitro Activities of Ceftaroline and Ceftriaxone Against *Streptococcus pneumoniae* Isolates With Various Resistance Profiles and by Specimen Source Collected in CANWARD, 2009 – 2010

S. pneumoniae (n) (by resistance profile or specimen source)	MIC interpretation			MIC (µg/mL)		
	% Susceptible[a]	% Intermediate	% Resistant	MIC_{50}	MIC_{90}	Range
All (420)						
Ceftaroline	100	0	0	≤0.03	≤0.03	≤0.03-0.25
Ceftriaxone	99.1/97.9	0.7/1.2	0.2/1	≤0.12	≤0.12	≤0.12-4
Penicillin MIC ≤0.06 µg/mL (362)						
Ceftaroline	100	0	0	≤0.03	≤0.03	≤0.03-0.25
Ceftriaxone	100/100	0/0	0/0	≤0.12	≤0.12	≤0.12-0.25
Penicillin MIC ≥0.12 µg/mL (58)						
Ceftaroline	100	0	0	≤0.03	0.12	≤0.03-0.25
Ceftriaxone	93.1/84.5	5.2/8.6	1.7/6.9	≤0.12	1	≤0.12-4
Clarithromycin-resistant (68)						
Ceftaroline	100	0	0	≤0.03	0.12	≤0.03-0.25
Ceftriaxone	94.1/88.2	4.4/5.9	1.5/5.9	≤0.12	1	≤0.12-4
Multidrug-nonsusceptible[b] (32)						
Ceftaroline	100	0	0	0.06	0.12	≤0.03-0.25
Ceftriaxone	87.5/76.3	9.4/13.2	3.1/10.5	≤0.12	2	≤0.12-4
Blood isolates (218)						
Ceftaroline	100	0	0	≤0.03	≤0.03	≤0.03-0.25
Ceftriaxone	99.1/98.6	0.9/0.5	0/0.9	≤0.12	≤0.12	≤0.12-2
Respiratory isolates (202)						
Ceftaroline	100	0	0	≤0.03	≤0.03	≤0.03-0.25
Ceftriaxone	99/97	0.5/2	0.5/1	≤0.12	≤0.12	≤0.12-4

[a] Ceftriaxone CLSI M100-S21 nonmeningitis breakpoint (≤1 µg/mL: susceptible; 2 µg/mL: intermediate; ≥4 µg/mL: resistant) /ceftriaxone CLSI M100-S21 meningitis breakpoint (≤0.5 µg/mL: susceptible; 1 µg/mL: intermediate; ≥2 µg/mL: resistant). Ceftaroline MICs were interpreted with the FDA breakpoint (≤0.25 µg/mL: susceptible).
[b] Multidrug-nonsusceptible (penicillin MIC ≥0.12 µg/mL and clarithromycin MIC >0.5 µg/mL).
MIC = minimum inhibitory concentration.

Table 3. In vitro Activities of Ceftaroline and Ceftriaxone Against the Most Common *Streptococcus pneumoniae* Serotypes Collected in CANWARD, 2009 – 2010

S. pneumoniae serotype (n)	Ceftaroline			Ceftriaxone		
	MIC_{50} (µg/mL)	MIC_{90} (µg/mL)	% Susceptible[a]	MIC_{50} (µg/mL)	MIC_{90} (µg/mL)	% Susceptible[b]
All (323)	≤0.03	≤0.03	100	≤0.12	≤0.12	98.8/97.5
19A (44)	≤0.03	0.12	100	≤0.12	1	90.9/88.6
22F (28)	≤0.03	≤0.03	100	≤0.12	≤0.12	100/100
3 (24)	≤0.03	≤0.03	100	≤0.12	≤0.12	100/100
4 (19)	≤0.03	≤0.03	100	≤0.12	≤0.12	100/100
7F (18)	≤0.03	≤0.03	100	≤0.12	≤0.12	100/100
23A (18)	≤0.03	≤0.03	100	≤0.12	≤0.12	100/100
6C (16)	≤0.03	0.06	100	≤0.12	≤0.12	100/100
19F (13)	≤0.03	0.12	100	≤0.12	0.25	100/92.3
12F (11)	≤0.03	≤0.03	100	≤0.12	≤0.12	100/100
Other (132)	≤0.03	≤0.03	100	≤0.12	≤0.12	100/98.5

[a] Ceftaroline MICs were interpreted with the FDA breakpoint (≤0.25 µg/mL: susceptible). [b]Ceftriaxone CLSI M100-S21 nonmeningitis breakpoint/ceftriaxone CLSI M100-S21 meningitis breakpoint.

Figure 1. MIC Distributions for Ceftaroline and Ceftriaxone Against All *Streptococcus pneumoniae* Collected in CANWARD, 2009 – 2010 (n=420)

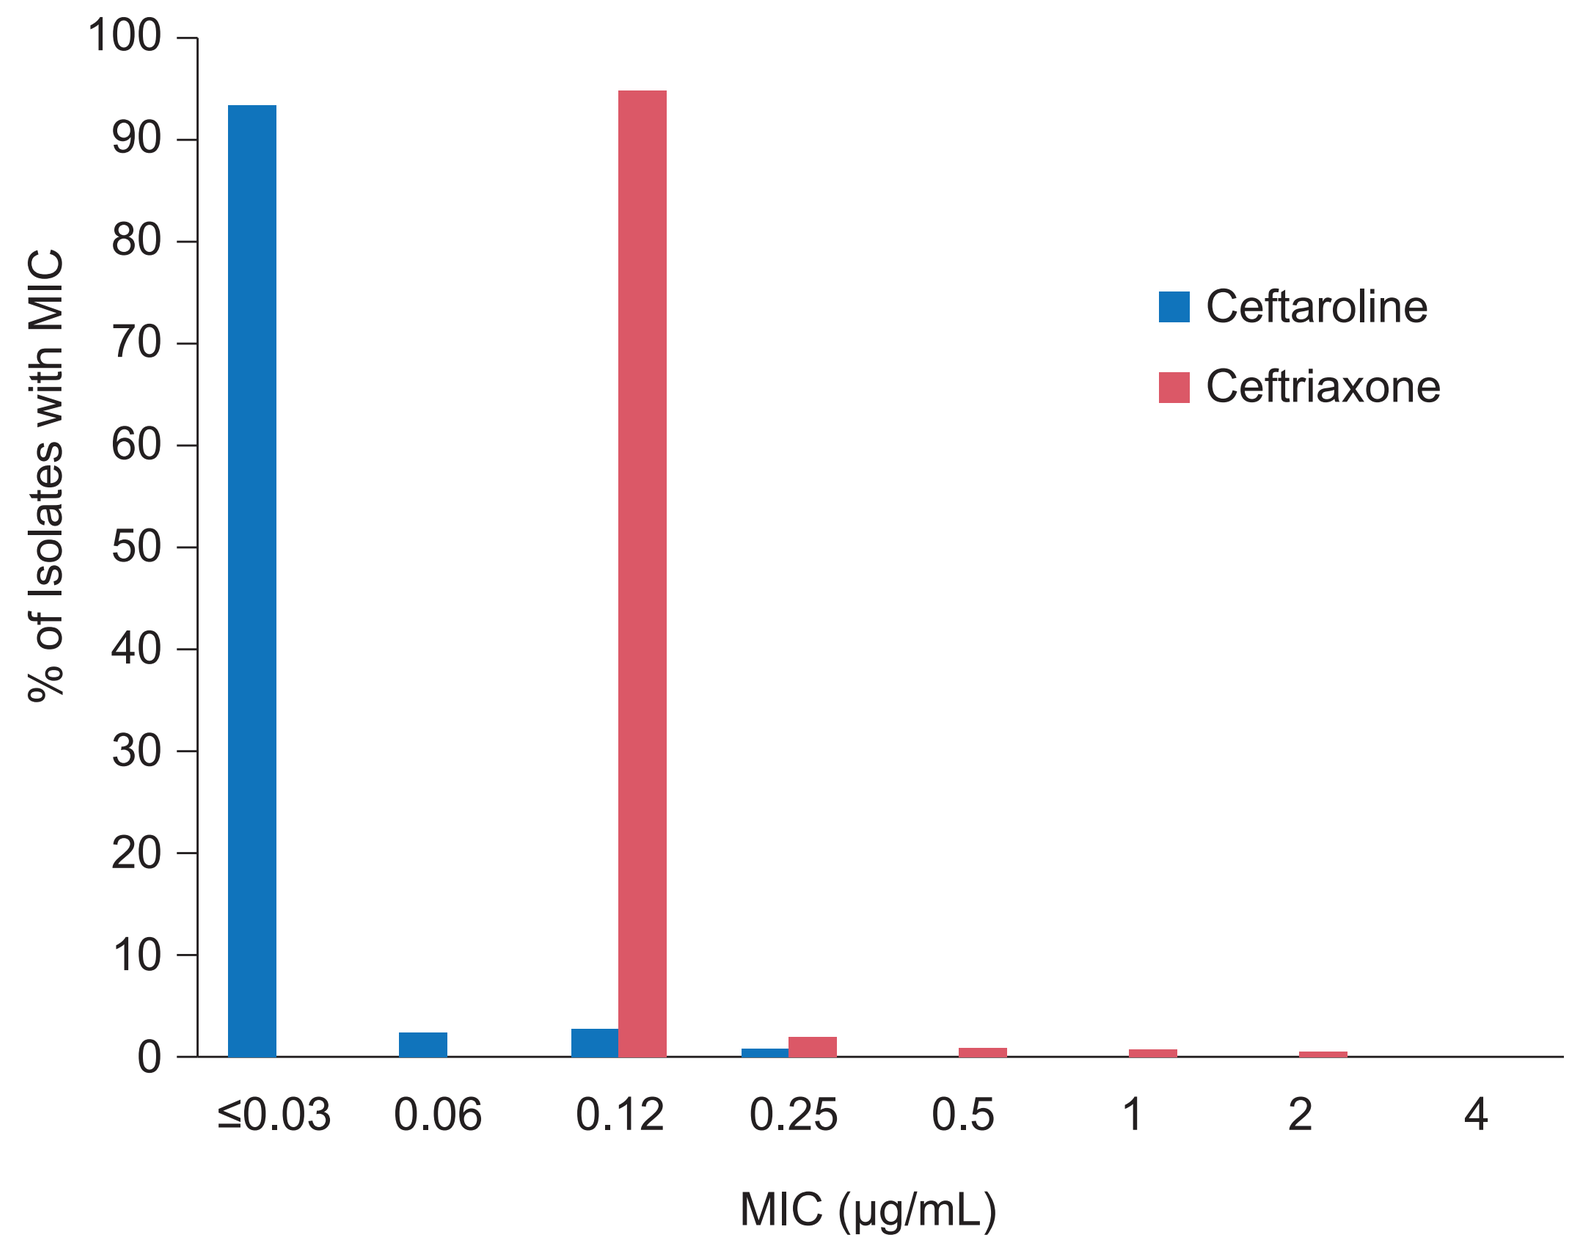
100
90
80
70
60
50
40
30
20
10
0
% of Isolates with MIC
Ceftaroline
Ceftriaxone
≤0.03
0.06
0.12
0.25
0.5
1
2
4
MIC (µg/mL)

Figure 2. MIC Distributions for Ceftaroline and Ceftriaxone Against *Streptococcus pneumoniae* With Penicillin MICs ≥0.12 µg/mL Collected in CANWARD, 2009 – 2010 (n=58)

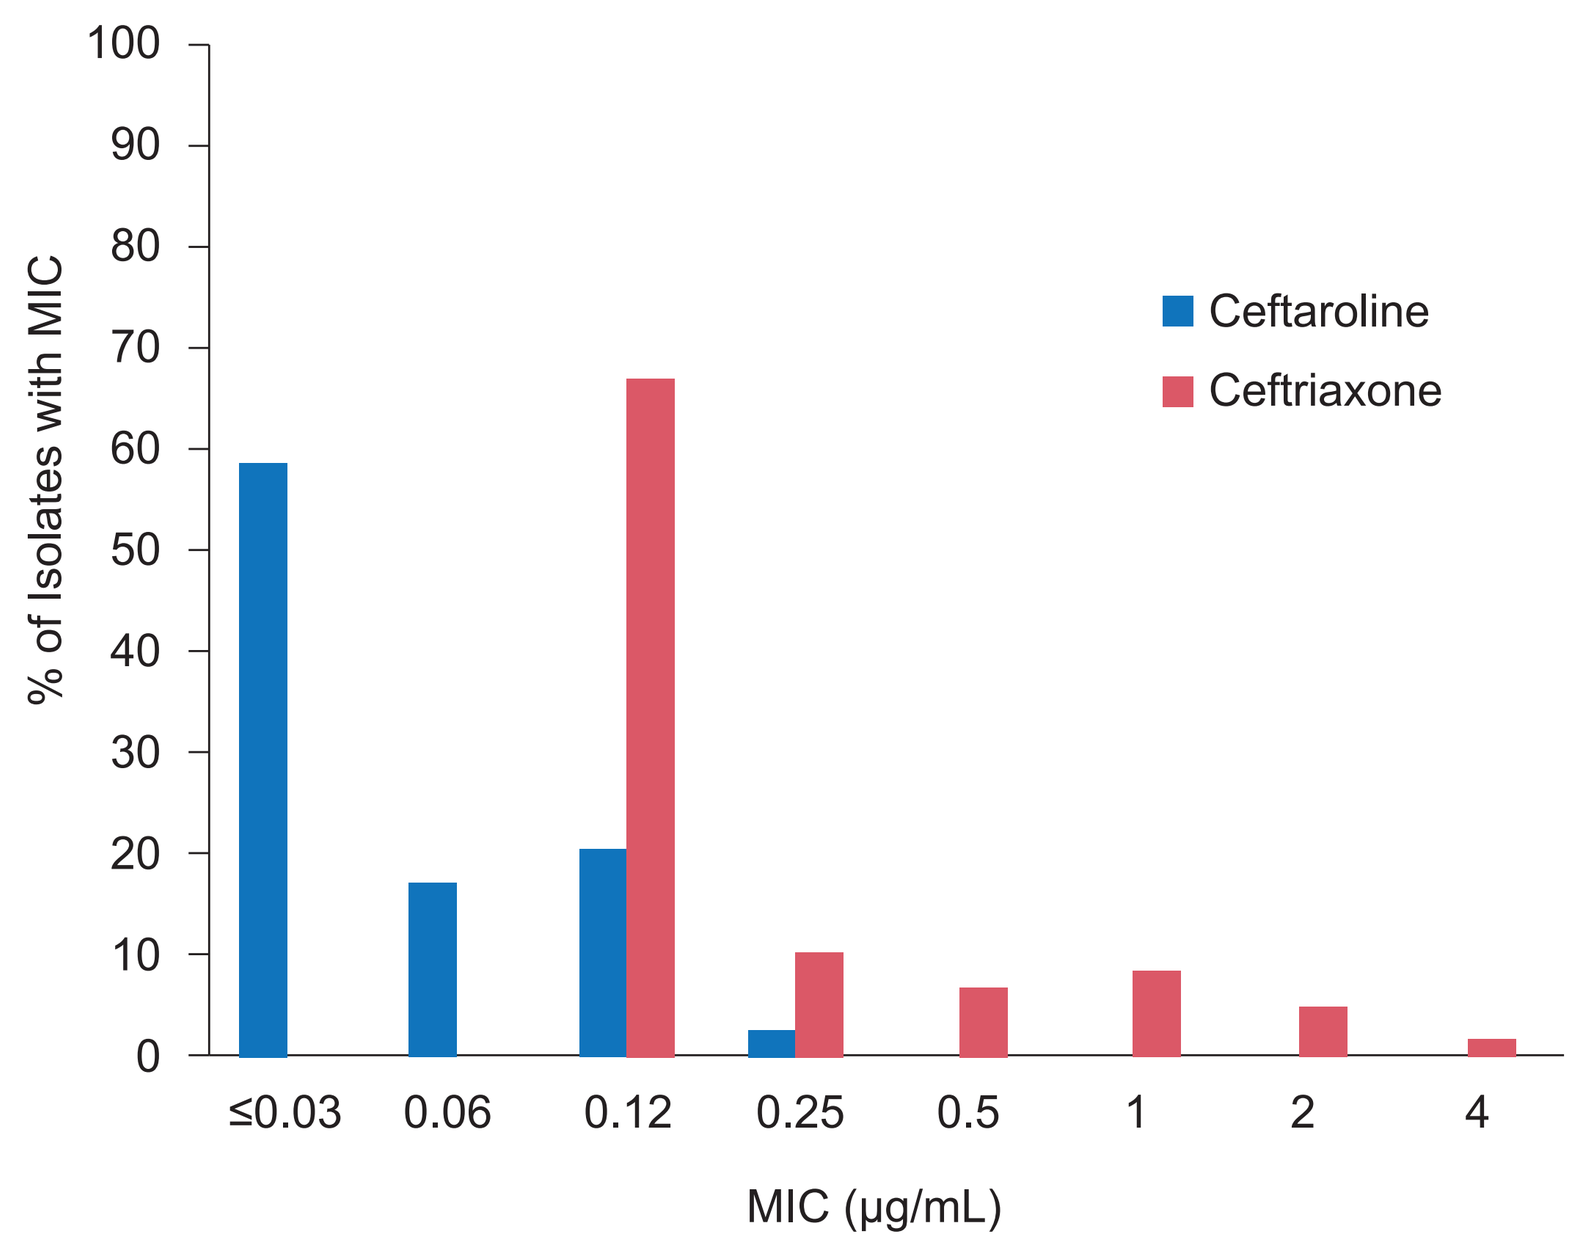
100
90
80
70
60
50
40
30
20
10
0
% of Isolates with MIC
Ceftaroline
Ceftriaxone
≤0.03
0.06
0.12
0.25
0.5
1
2
4
MIC (µg/mL)

Figure 3. MIC Distributions for Ceftaroline and Ceftriaxone Against Multidrug-Nonsusceptible *Streptococcus pneumoniae* (penicillin MIC ≥0.12 µg/mL and clarithromycin MIC >0.5 µg/mL) Collected in CANWARD, 2009 – 2010 (n=32)

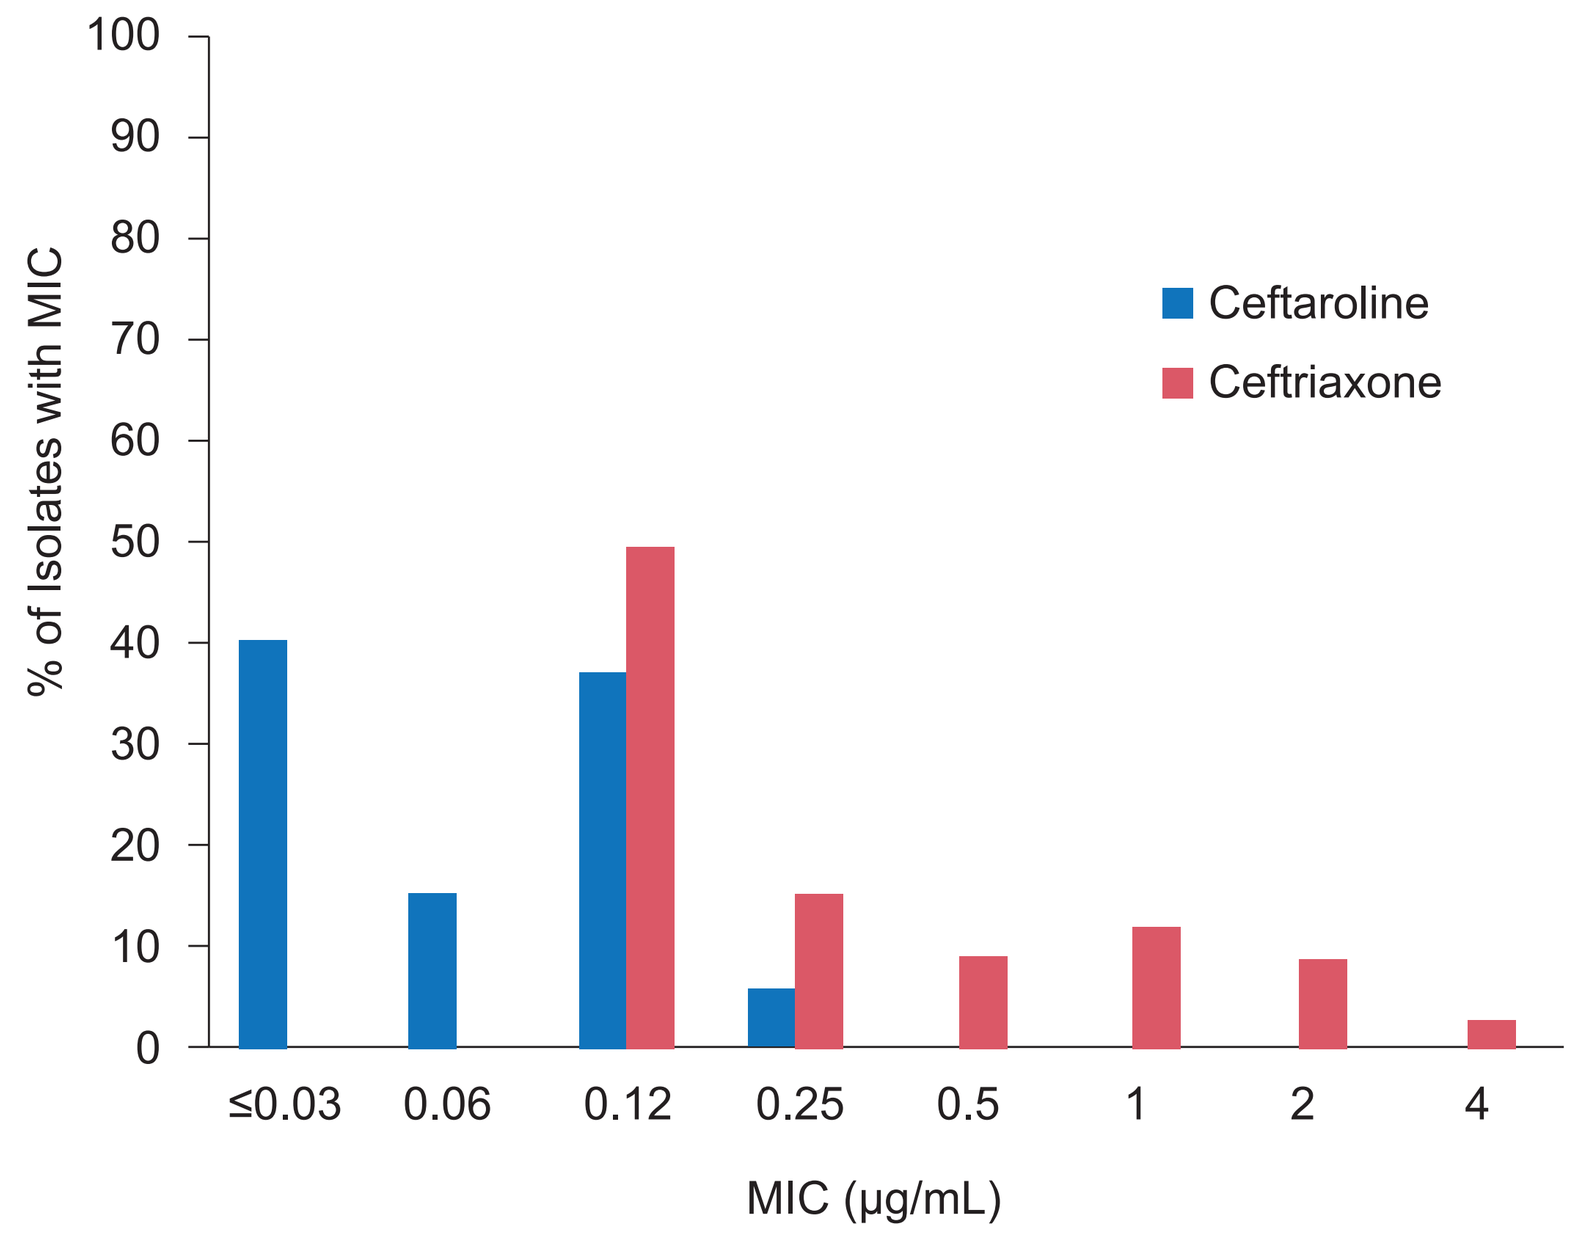
100
90
80
70
60
50
40
30
20
10
0
% of Isolates with MIC
Ceftaroline
Ceftriaxone
≤0.03
0.06
0.12
0.25
0.5
1
2
4
MIC (µg/mL)

Figure 4. MIC Distribution of Ceftaroline by *Streptococcus pneumoniae* Serotype (n=323)

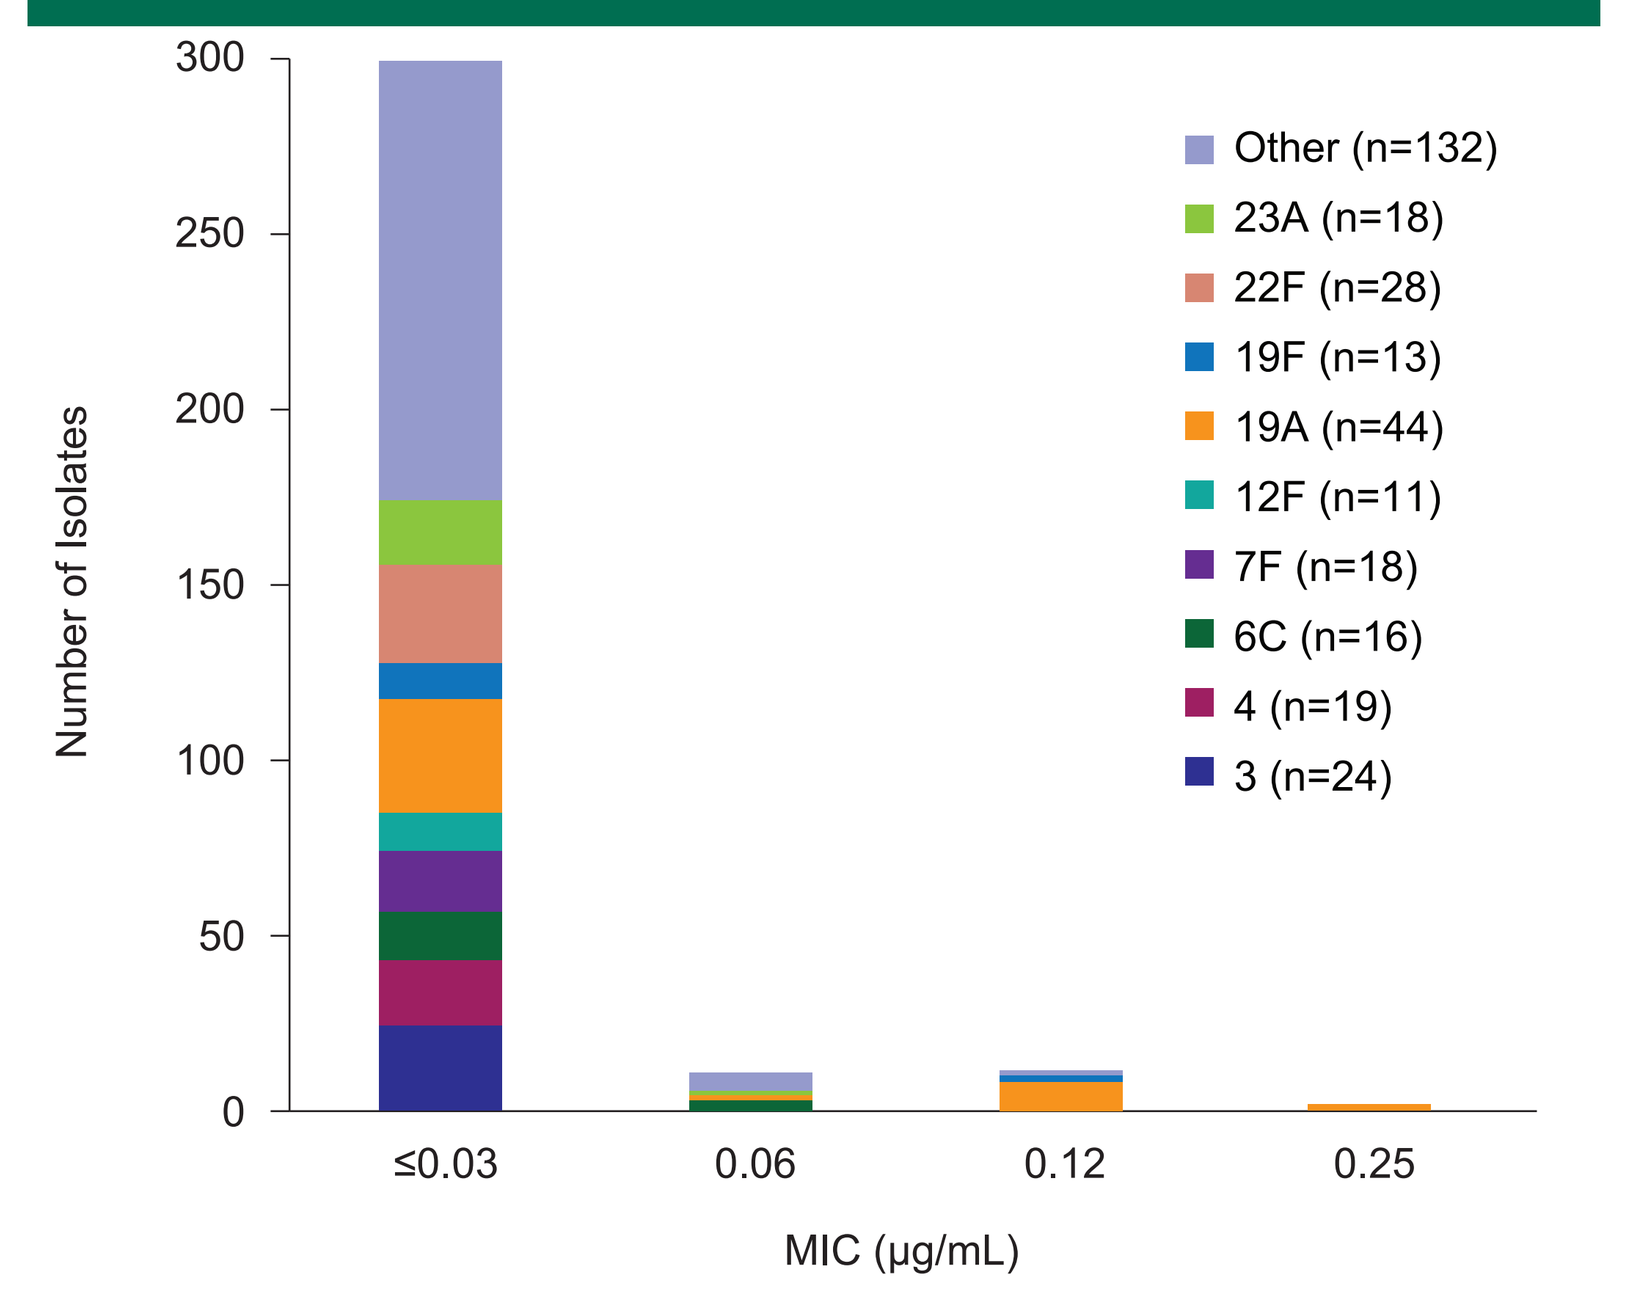
300
250
200
150
100
50
0
Number of Isolates
Other (n=132)
23A (n=18)
22F (n=28)
19F (n=13)
19A (n=44)
12F (n=11)
7F (n=18)
6C (n=16)
4 (n=19)
3 (n=24)
≤0.03
0.06
0.12
0.25
MIC (µg/mL)

Conclusions

- Ceftaroline demonstrated 8-fold greater in vitro activity than ceftriaxone against blood and respiratory *S. pneumoniae* isolates with penicillin MICs ≥0.12 µg/mL and against clarithromycin-resistant isolates.
- Ceftaroline demonstrated 16-fold greater in vitro activity than ceftriaxone against multidrug-nonsusceptible *S. pneumoniae* isolated from blood cultures and respiratory specimens.
- Ceftaroline remains highly active against emerging *S. pneumoniae* serotypes. Ceftaroline was 8-fold more active than ceftriaxone against serotype 19A isolates.
- Ceftaroline fosamil has the potential to be a useful agent in the treatment of *S. pneumoniae* infections, including penicillin- and multidrug-nonsusceptible strains.

References

1. Zhanel GG, DeCorby M, Adam H, et al, the Canadian Antimicrobial Resistance Alliance, Hoban DJ. Prevalence of antimicrobial-resistant pathogens in Canadian hospitals: results of the Canadian Ward Surveillance Study (CANWARD 2008). *Antimicrob Agents Chemother*. 2010;54:4684-4693.
2. Zhanel GG, Sniezek G, Schweizer F, et al. Ceftaroline: a novel broad-spectrum cephalosporin with activity against methicillin-resistant *Staphylococcus aureus*. *Drugs*. 2009;69:809-831.
3. Moisan H, Pruneau M, Malouin F. Binding of ceftaroline to penicillin-binding proteins of *Staphylococcus aureus* and *Streptococcus pneumoniae*. *J Antimicrob Chemother*. 2010;65:713-716.
4. Low DE, File TM Jr, Eckburg PB, et al. FOCUS 2: a randomized, double-blinded, multicenter, Phase III trial of the efficacy and safety of ceftaroline fosamil versus ceftriaxone in community-acquired pneumonia. *J Antimicrob Chemother*. 2011;66:iii33-iii44.
5. Clinical and Laboratory Standards Institute. *Performance Standards for Antimicrobial Susceptibility Testing: Twenty-First Informational Supplement. M100-S21.* 2011. Wayne, PA: Clinical and Laboratory Standards Institute.
6. Austrian R. The quellung reaction, a neglected microbiologic technique. *Mt Sinai J Med*. 1976;43:699-709.

Acknowledgments

This study was supported in part by the University of Manitoba and Forest Laboratories, Inc.

The contributions of the CANWARD study sites and investigators are gratefully acknowledged: Queen Elizabeth II HSC, Nova Scotia (Dr R Davidson); South East Regional Health Authority, New Brunswick (Dr M Kuhn); Hopital Maisonneuve-Rosemont, Quebec (Dr M Laverdiere); Montreal General Hospital, Quebec (Dr V Loo); Royal Victoria Hospital, Quebec (Dr V Loo); Centre Hospitalier Regional de Trois-Rivieres, Quebec (Dr M Goyette); Children's Hospital of Eastern Ontario, Ontario (Dr F Chan); London Health Sciences Centre, Ontario (Dr Z Hussain); The Ottawa Hospital, Ontario (Dr M Desjardins) (2009 only); Mount Sinai Hospital, Ontario (Dr S Poutanen); St. Joseph's Hospital, Ontario (Dr C Lee); St. Michael's Hospital, Ontario (Dr L Matukas); Health Sciences Centre, Manitoba (Drs D Hoban/G Zhanel); Royal University Hospital, Saskatchewan (Dr J Blondeau); University of Alberta Hospital, Alberta (Dr R Rennie); Vancouver Hospital, British Columbia (Dr D Roscoe).

Forest Laboratories, Inc., was involved in the decision to present these results. Forest Laboratories, Inc., had no involvement in the design, collection, analysis, or interpretation of data.

Scientific Therapeutics Information, Inc, provided editorial coordination and production services, which were funded by Forest Research Institute, Inc.